Poster 1300

Efficacy and Safety of Daclatasvir in Combination With Asunaprevir in Cirrhotic and Non-Cirrhotic Patients With HCV Genotype 1b: Results of the HALLMARK-DUAL Study

Kao J-H,[1] Heo J,[2] Yoffe B,[3] Sievert W,[4] Jacobson IM,[5] Bessone F,[6] Peng C-Y,[7] Roberts S,[8] Yoon KT,[9] Kopit J,[10] Linaberry M,[11] Noviello S,[11] Hughes E,[11] Manns M[12]

[1]National Taiwan University Hospital, Taipei, Taiwan; [2]Pusan National University Hospital, Busan, Republic of Korea; [3]VAMC, Baylor College of Medicine, Houston, Texas, USA; [4]Monash Health and Monash University, Melbourne, Australia; [5]Weill Cornell Medical College, New York, New York, USA; [6]Instituto CAICI, Rosario, Argentina; [7]School of Medicine, China Medical University, Taichung, Taiwan; [8]The Alfred Hospital, Melbourne, Australia; [9]Pusan National University Yangsan Hospital, Busan, Republic of Korea; [10]Bristol-Myers Squibb Research and Development, Wallingford, Connecticut, USA; [11]Bristol-Myers Squibb Research and Development, Princeton, New Jersey, USA; [12]Hannover Medical School, Hannover, Germany

Corresponding author:
Jia-Horng Kao (kaojh@ntu.edu.tw)

BACKGROUND

- Current treatment of chronic HCV infection in patients with cirrhosis is complicated by poor tolerability and suboptimal sustained virologic response (SVR) rates with peginterferon/ribavirin (pegIFN/RBV) alone or in combination with direct-acting antivirals[1–3]
- In a phase 3 Japanese study in genotype (GT) 1b (AI447-026), all-oral daclatasvir (DCV) plus asunaprevir (ASV) demonstrated high SVR_{24} rates in IFN-nonresponders and ineligible-naive/intolerant patients with and without cirrhosis (91% and 84%, respectively)[4]
- DCV + ASV was evaluated in a global phase 3 study in patients with GT 1b who were treatment-naive, nonresponders to pegIFN/RBV, or pegIFN/RBV ineligible/intolerant
 - Daclatasvir (DCV)
 - Potent, pangenotypic[a] NS5A inhibitor
 - Once daily with low potential for drug–drug interactions[b]
 - Safe and well tolerated in > 5500 patients treated
 - Asunaprevir (ASV)
 - NS3 protease inhibitor
 - Clinical data in GT 1 and 4
 - Generally well tolerated in > 2000 patients treated
- Primary efficacy, safety, and resistance results from this study are presented separately (Late Breakers session, oral 0166)[5]
- Efficacy and safety results in patients with and without cirrhosis are presented here

[a]Pangenotypic: GT 1–6 *in vitro* and GT 1–4 in clinical trials.
[b]No clinically significant drug-drug interactions with antiretrovirals and transplant medications.

METHODS

- This global phase 3 study evaluated DCV (60-mg tablet once daily) combined with ASV (100-mg softgel capsule twice daily) in patients with GT 1b

Figure 1. HALLMARK-DUAL Study (AI447-028) Design

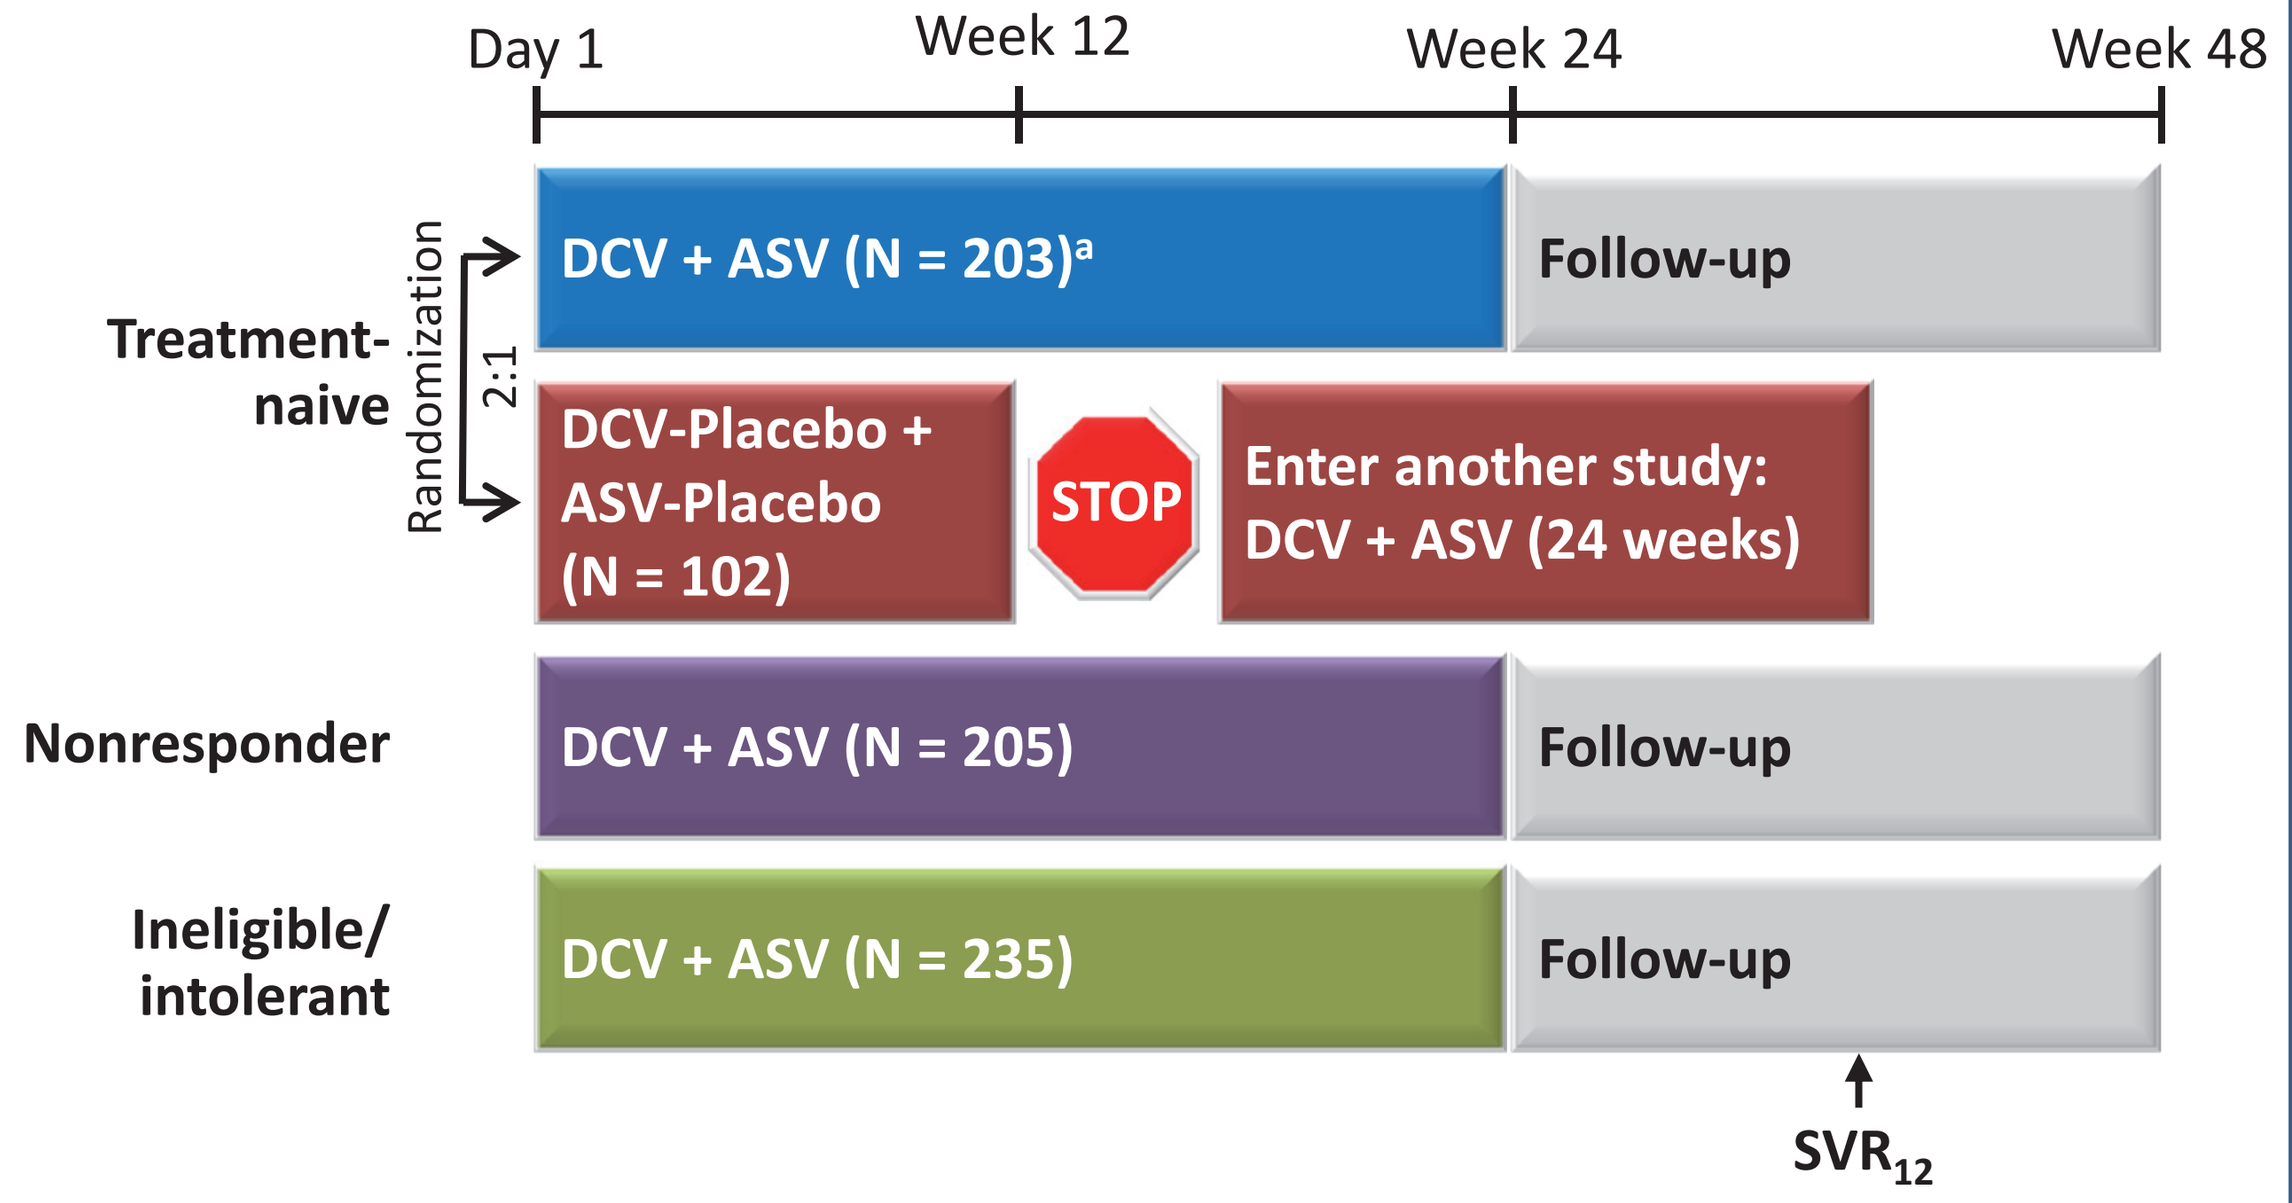
Day 1
Week 12
Week 24
Week 48
Treatment-naive
Randomization
2:1
DCV + ASV (N = 203)a
Follow-up
DCV-Placebo + ASV-Placebo (N = 102)
STOP
Enter another study: DCV + ASV (24 weeks)
Nonresponder
DCV + ASV (N = 205)
Follow-up
Ineligible/intolerant
DCV + ASV (N = 235)
Follow-up
SVR12

[a]Excludes 2 patients inadvertently assigned, instead of randomized, to DCV + ASV; patients were excluded from efficacy analyses but both achieved SVR_{12}.

METHODS (cont)

- Patients: men and women aged ≥ 18 years
 - Treatment-naive
 - Prior pegIFN/RBV nonresponders (null or partial response)
 - PegIFN/RBV ineligible/intolerant (treatment-naive or -experienced)
 - Depression
 - Anemia/neutropenia
 - Compensated advanced fibrosis/cirrhosis (F3/F4) with thrombocytopenia (screening platelets 50 to < 90 x 10^9 cells/L and/or history of thrombocytopenia on pegIFN/RBV)
- Primary endpoint: proportion of DCV + ASV-treated patients with SVR_{12}
- Efficacy and safety with DCV + ASV were assessed in patients with and without cirrhosis
 - Presence of cirrhosis assessed by liver biopsy or transient elastography (FibroScan; ≥ 14.6 kPa)

RESULTS

Table 1. Demographic and Baseline Disease Characteristics

Parameter	Cirrhotic (N = 223)	Non-cirrhotic (N = 524)
Age, median years	59	56
Male, n (%)	121 (54)	243 (46)
Race, n (%)		
White	153 (69)	358 (68)
Black	8 (4)	34 (6)
Asian	60 (27)	126 (24)
Other[a]	2 (1)	6 (1)
HCV RNA, n (%)		
< 800,000 log_{10} IU/mL	48 (22)	106 (20)
≥ 800,000 log_{10} IU/mL	175 (78)	418 (80)
IL28B genotype, n (%)		
CC	65 (33)	122 (28)
Non-CC	132 (67)	317 (72)
Prior treatment experience, n (%)		
Treatment naive[b]	49 (22)	258 (49)
Nonresponder	63 (28)	142 (27)
Ineligible/intolerant[c]	111 (50)	124 (24)

[a]Includes North African, American Indian/Alaska Native, Native Hawaiian/other Pacific islander, Afghanistan (Middle East), mixed, and southwest Asian.
[b]Cirrhotic: 33 DCV + ASV, 16 placebo; non-cirrhotic: 172 DCV + ASV, 86 placebo.
[c]Prior anti-HCV treatment taken by 182/235 (77%) patients.

- Baseline characteristics were comparable in cirrhotic and non-cirrhotic patients
- 90% of cirrhotic patients and 89% of non-cirrhotic patients completed DCV + ASV treatment

- SVR_{12} was achieved in 91% of treatment-naive patients, 82% of nonresponders, and 83% of ineligible/intolerant patients[5,a]
 - SVR_{12} rates were 90%, 82%, and 82%, respectively, when patients with missing data were counted as treatment failures

[a]SVR_{12} documented on or after posttreatment Week 12.

RESULTS (cont)

Figure 2. Virologic Response by Cirrhosis Status

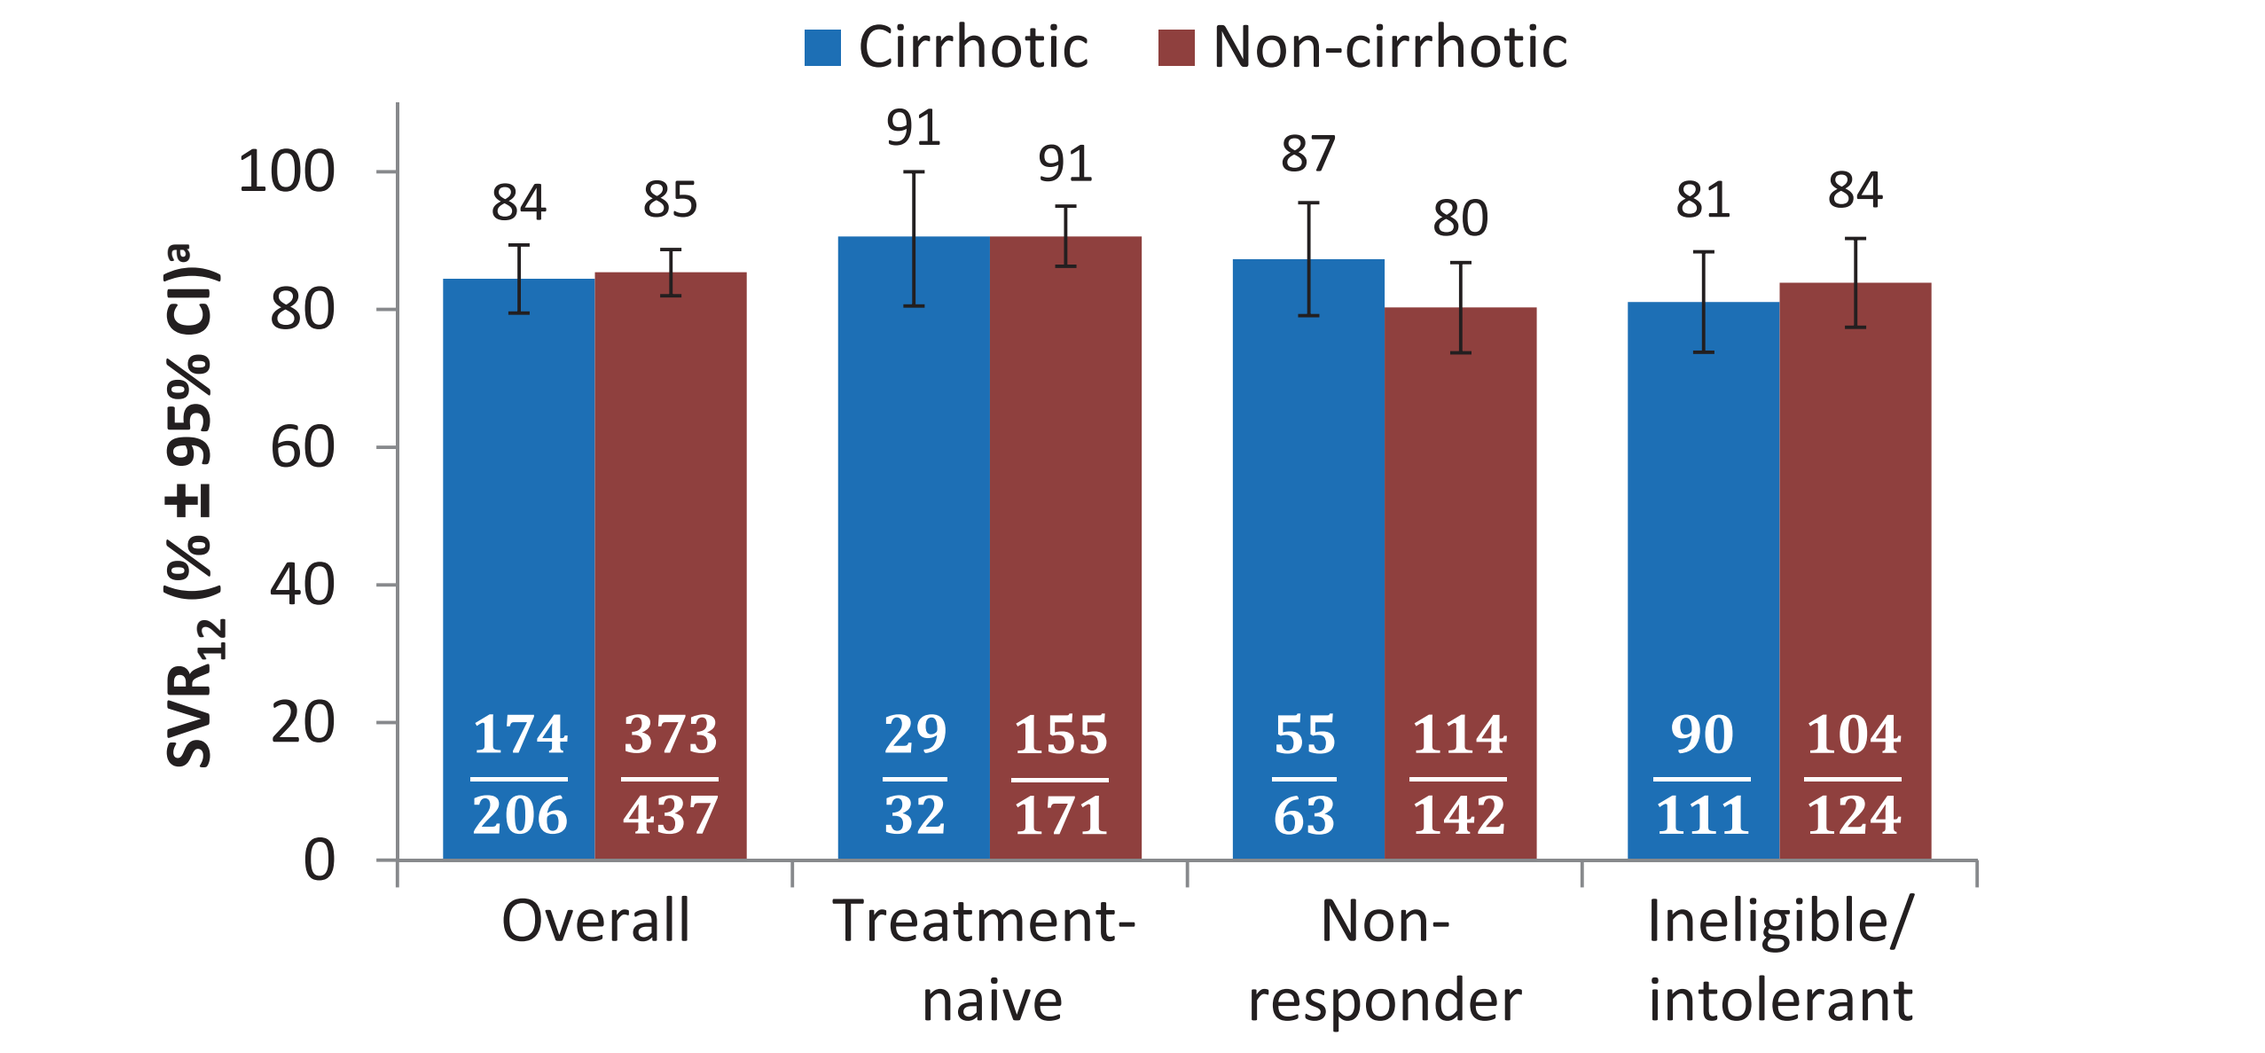
Cirrhotic
Non-cirrhotic
SVR12 (% ± 95% CI)a
100
80
60
40
20
0
84 85 91 91 87 80 81 84
174/206 373/437
29/32 155/171
55/63 114/142
90/111 104/124
Overall
Treatment-naive
Non-responder
Ineligible/intolerant

- SVR_{12} was achieved in a similar proportion of cirrhotic and non-cirrhotic patients
- When patients with missing SVR_{12} data were counted as treatment failures, rates were identical except for treatment-naive non-cirrhotic patients (89%) and ineligible/intolerant cirrhotic patients (79%)
- 69% of cirrhotic patients and 76% of non-cirrhotic patients had undetectable HCV RNA at on-treatment Week 4

[a]SVR_{12} documented on or after posttreatment Week 12.

Figure 3. Virologic Response in Cirrhotics and Non-Cirrhotics by Patient Subgroup

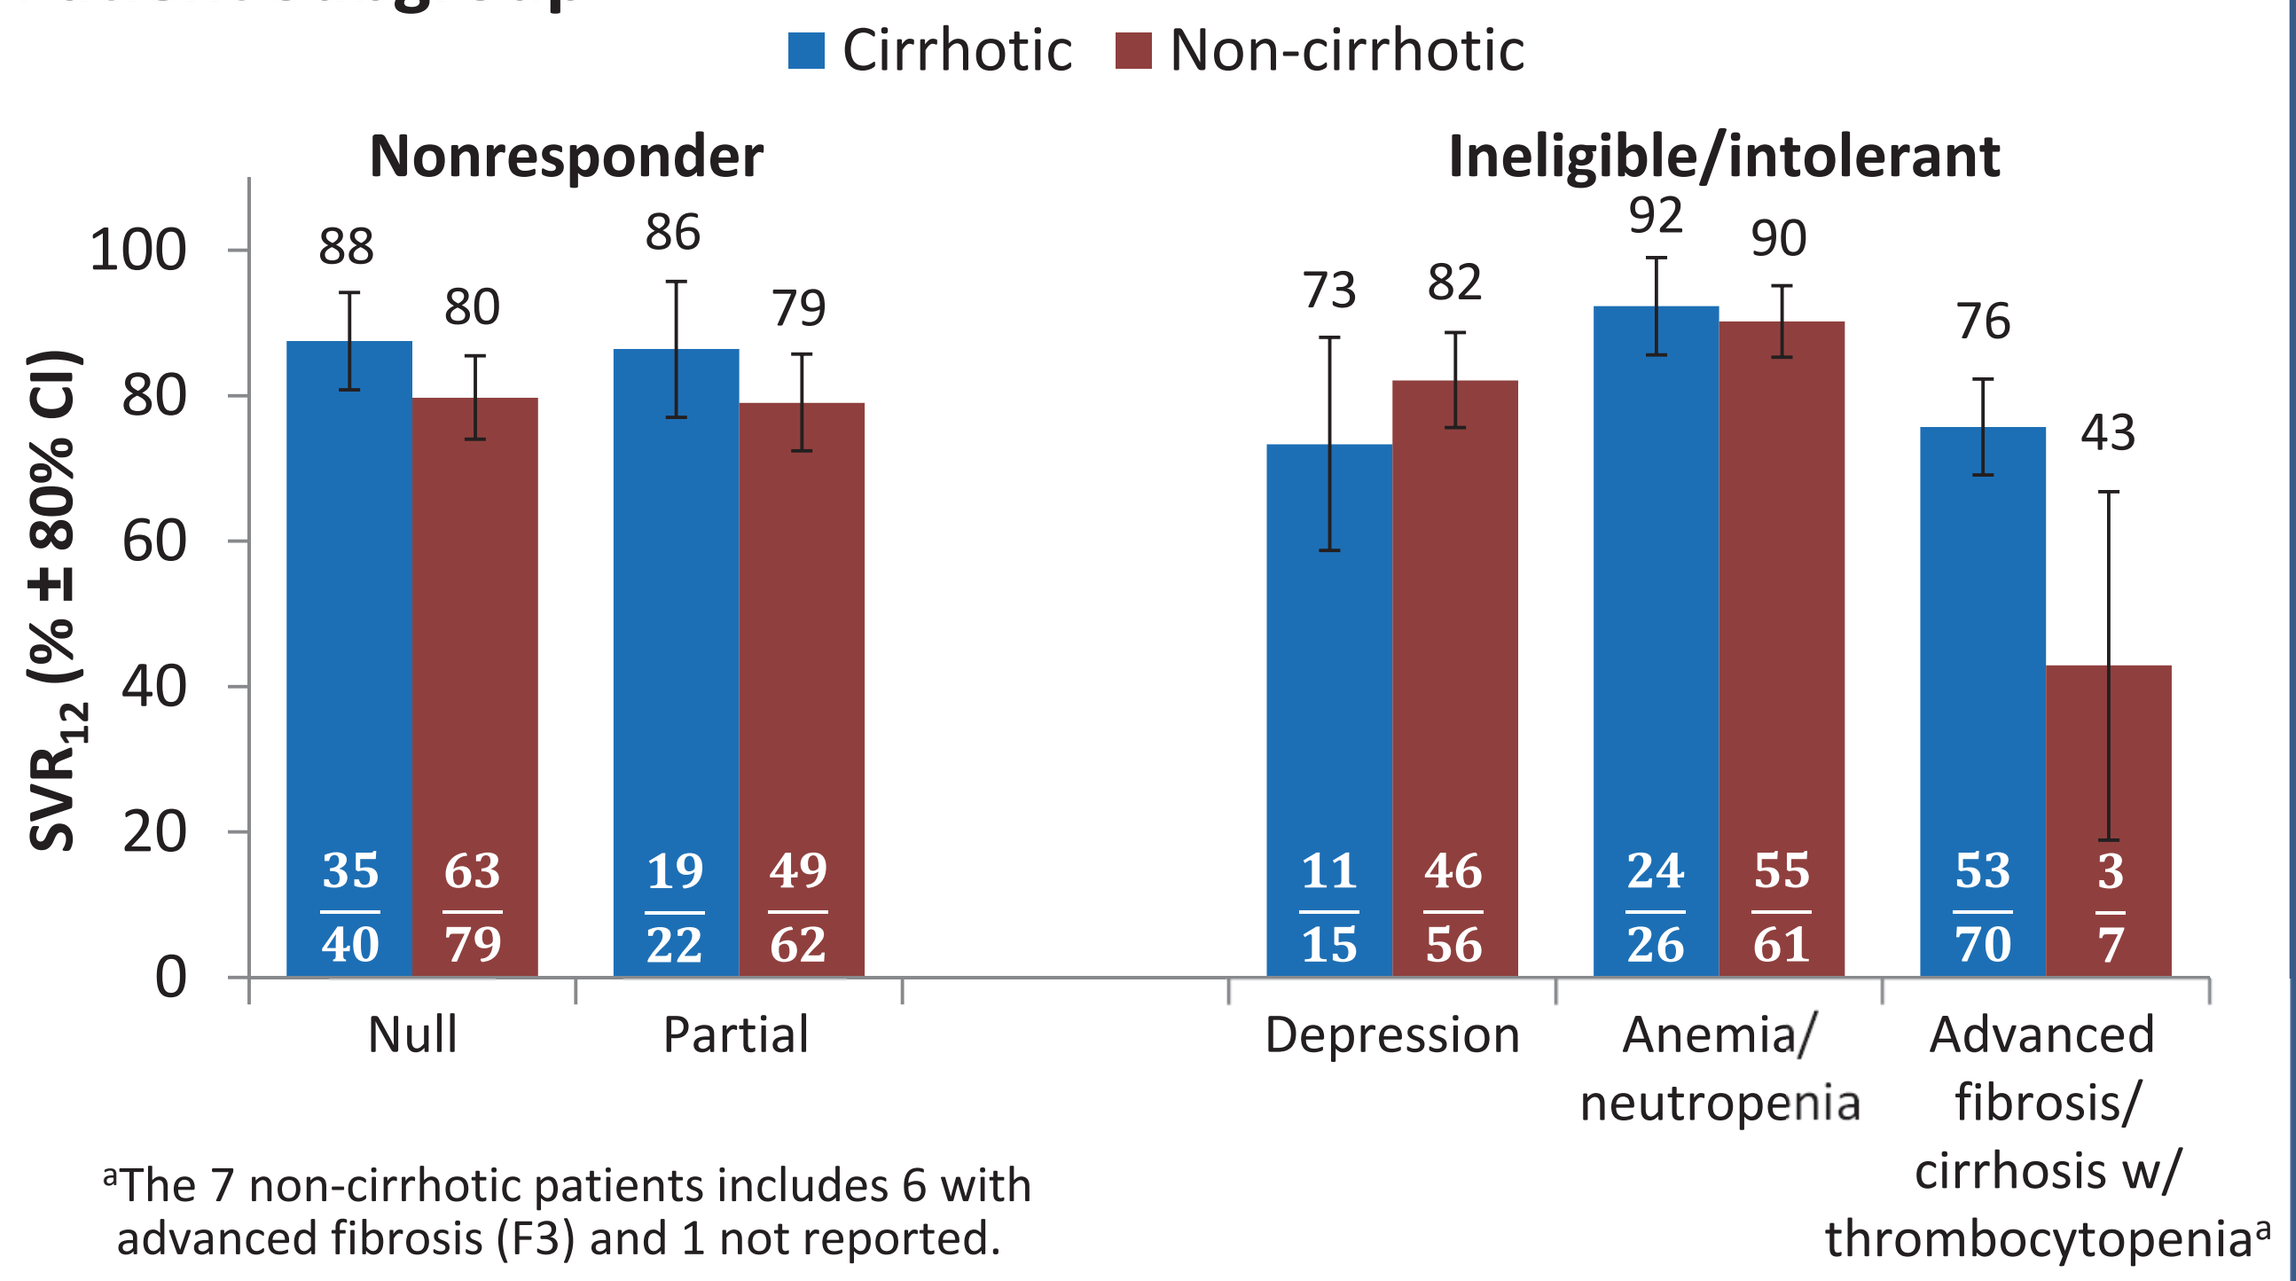
Cirrhotic
Non-cirrhotic
Nonresponder
Ineligible/intolerant
SVR12 (% ± 80% CI)
100
80
60
40
20
0
88 80 86 79 73 82 92 90 76 43
35/40 63/79
19/22 49/62
11/15 46/56
24/26 55/61
53/70 3/7
Null
Partial
Depression
Anemia/neutropenia
Advanced fibrosis/cirrhosis w/ thrombocytopeniaa

[a]The 7 non-cirrhotic patients includes 6 with advanced fibrosis (F3) and 1 not reported.

- Patients with cirrhosis achieved high SVR_{12} rates across all subgroups
- Among ineligible/intolerant patients, SVR_{12} rate was slightly lower in those with advanced fibrosis/cirrhosis with thrombocytopenia (73%) versus depression (80%) and anemia/neutropenia (91%)

REFERENCES

1. Bourlière M, *et al. Liver Int* 2012;32(Suppl 1):113–119.
2. Lawitz E, *et al. N Engl J Med* 2013;368:1878–1887.
3. OLYSIO™ (simeprevir) prescribing information. Food and Drug Administration, 2014. (http://www.accessdata.fda.gov/drugsatfda_docs/label/2013/205123s001lbl.pdf.)
4. Kumada H, *et al. Hepatology* 2014 (ePub). doi:10.1002/hep.27113.
5. Manns M *et al.* 49th *EASL*; Apr 9–13, 2014; London, United Kingdom. Late Breakers oral 0166.

RESULTS (cont)

Figure 4. Virologic Response by Baseline Platelet Count

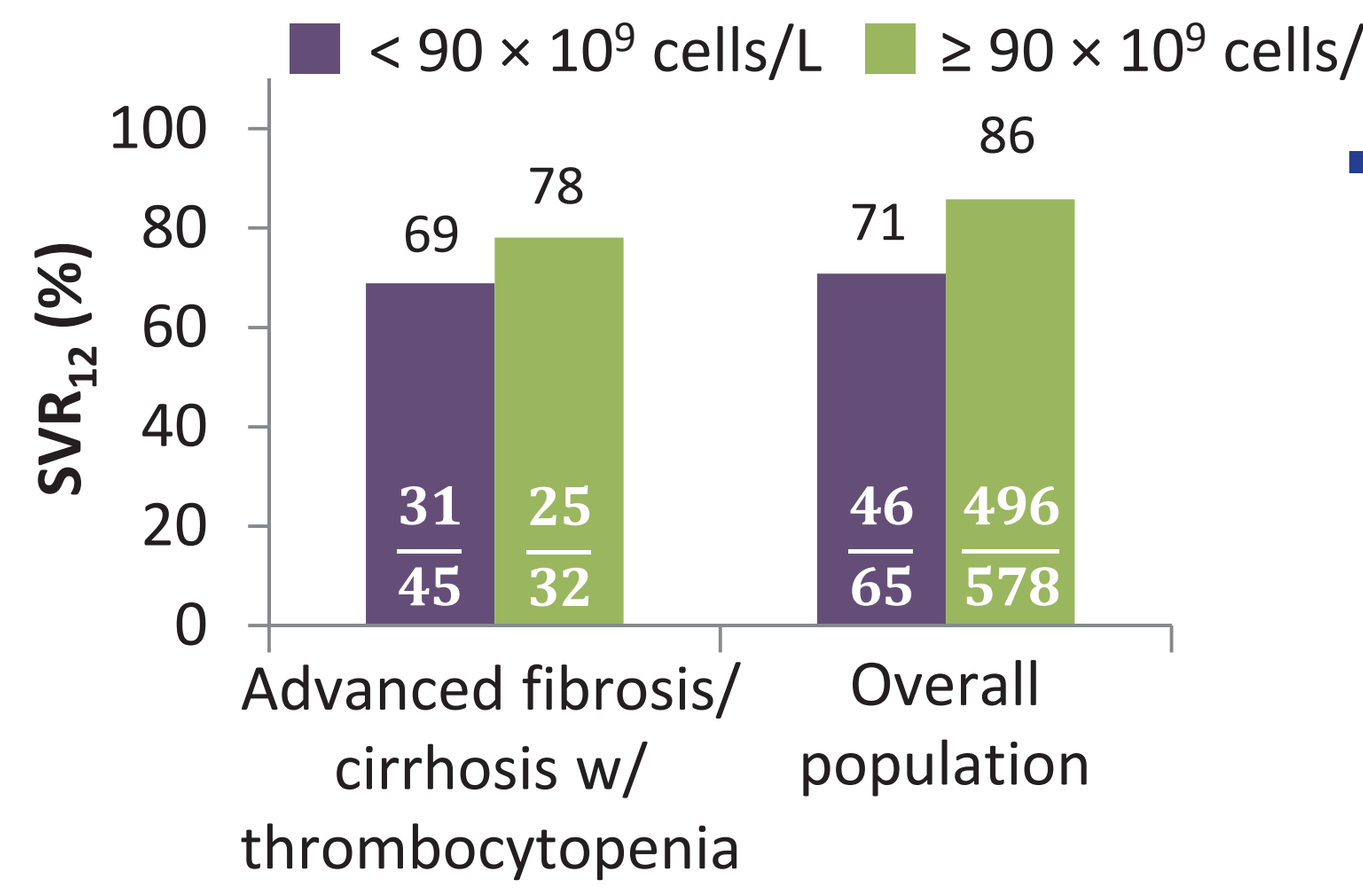
< 90 × 10^9 cells/L
≥ 90 × 10^9 cells/L
SVR12 (%)
100
80
60
40
20
0
69 78 71 86
31/45 25/32
46/65 496/578
Advanced fibrosis/cirrhosis w/ thrombocytopenia
Overall population

- In the subgroup with advanced fibrosis/cirrhosis with thrombocytopenia, SVR_{12} rates were lower in patients with baseline platelet counts < 90 versus ≥ 90 x 10^9 cells/L
 - Similar results seen in the overall patient population

Figure 5. Virologic Response in Cirrhotics by Baseline Factors

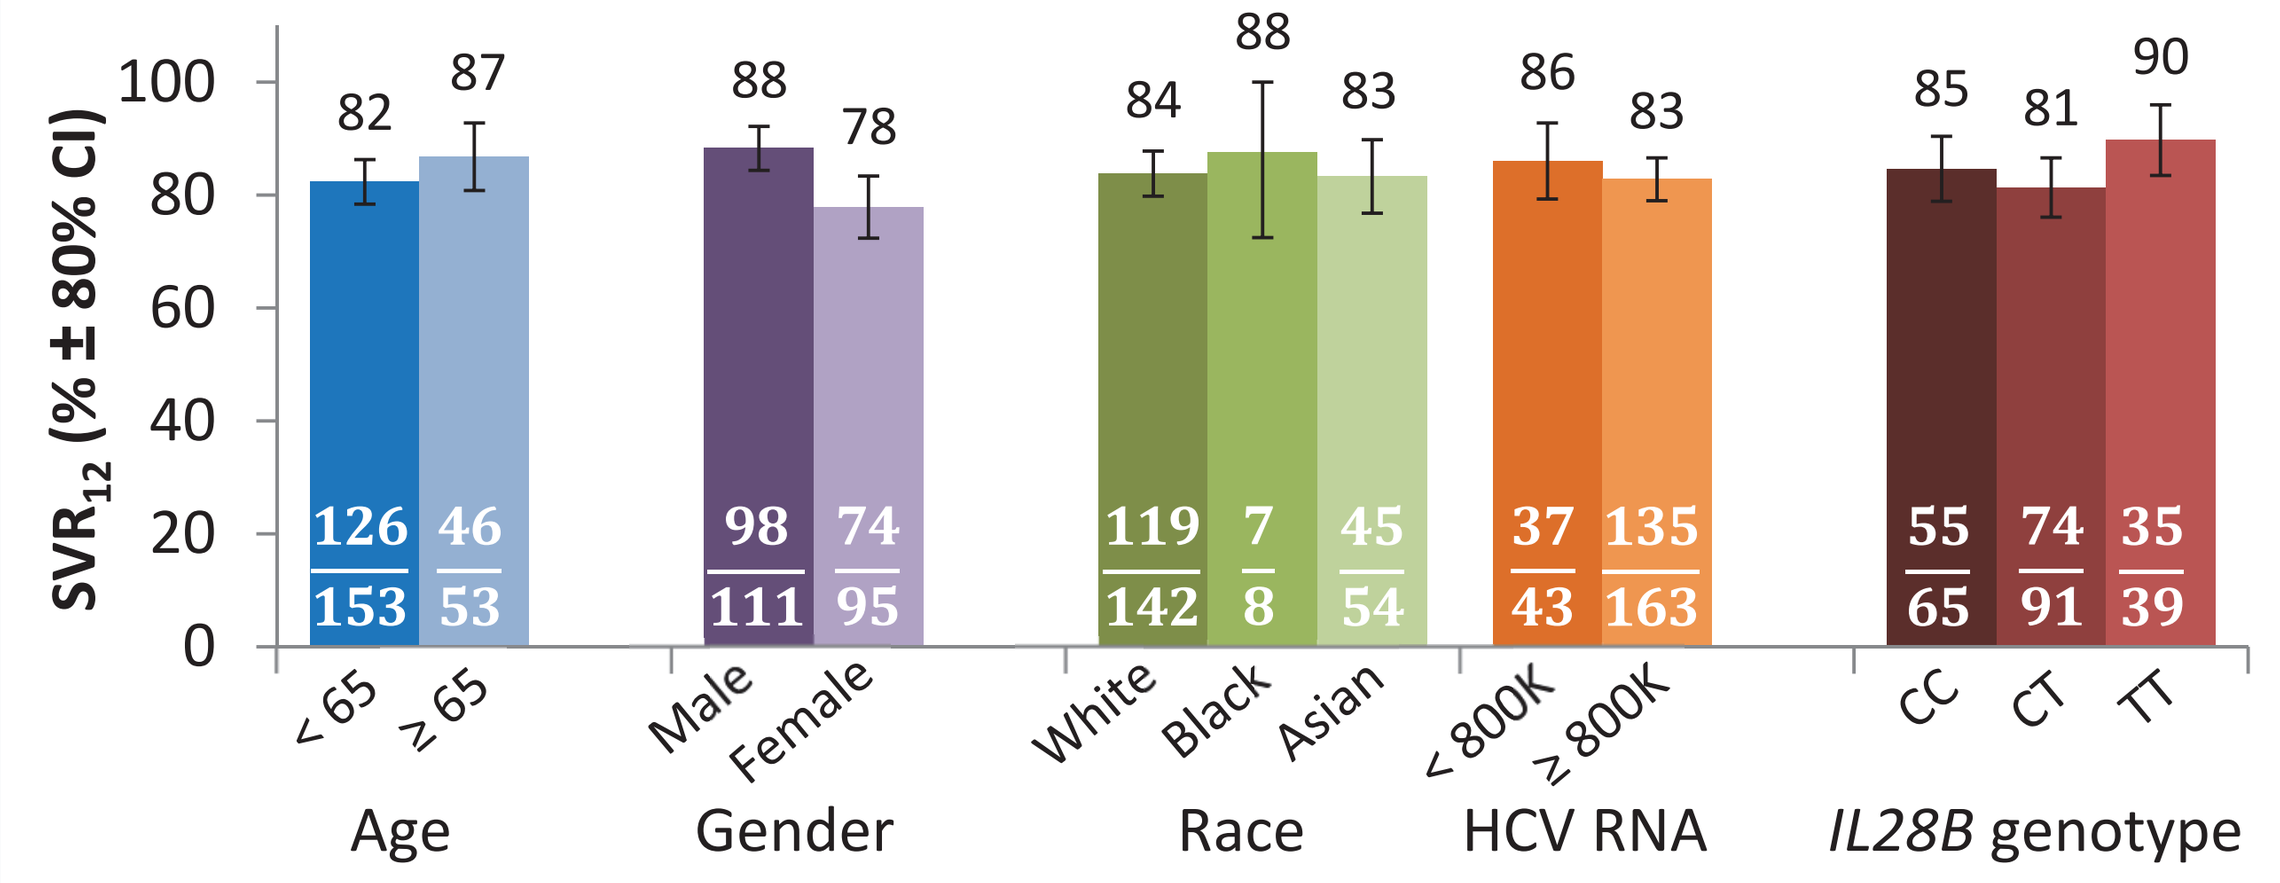
SVR12 (% ± 80% CI)
100
80
60
40
20
0
82 87 88 78 84 88 83 86 83 85 81 90
126/153 46/53
98/111 74/95
119/142 7/8 45/54
37/43 135/163
55/65 74/91 35/39
< 65
≥ 65
Male
Female
White
Black
Asian
< 800K
≥ 800K
CC
CT
TT
Age
Gender
Race
HCV RNA
IL28B genotype

- Cirrhotic and non-cirrhotic patients had similar virologic responses based on baseline factors

Table 2. Patients Without SVR_{12} by Cirrhosis Status

Patients, n (%)	Cirrhotic (N = 206)	Non-cirrhotic (N = 437)
All	34 (17)	67 (15)
On-treatment failures		
Virologic breakthrough	16 (8)	39 (9)
Detectable or missing RNA at end of treatment	8 (4)	8 (2)
Posttreatment failures		
Relapse[a]	8 (5)	16 (4)
Missing RNA at posttreatment Week 12[a]	2 (1)	4 (1)

[a]Percentages based on number of patients with undetectable HCV RNA at end of treatment (cirrhotic, n = 175; non-cirrhotic, n = 373).

- Similar proportions of cirrhotic and non-cirrhotic patients did not achieve SVR_{12}
- 14 of 28 cirrhotic patients with baseline NS5A L31 and/or Y93 resistance-associated variants achieved SVR_{12}[a]

[a]Data on resistance in overall study reported in Late Breakers session oral 166.[5]

ACKNOWLEDGMENTS & DISCLOSURES

- The authors thank the patients, their families, and staff at all study sites
- The authors thank M Lovegren, G Denisky, and M Mohebbian for their contributions to study execution
- ClinicalTrials.gov, registration number NCT01581203 (Study AI447-028)
- JH Kao has no conflicts to disclose
- J Kopit, M Linaberry, S Noviello, and E Hughes are employees of Bristol-Myers Squibb
- Editorial support was provided by J Loh of Articulate Science and funded by Bristol-Myers Squibb

RESULTS (cont)

Table 3. On-Treatment Safety and Tolerability by Cirrhosis Status

Patients, n (%)	Cirrhotic (N = 207)[a]	Non-cirrhotic (N = 438)[a]
Death	0	0
Serious AEs	13 (6)	26 (6)
AEs leading to discontinuation[b]	1 (0.5)	9 (2)
Common AEs (≥ 10% of patients)		
Headache	51 (25)	108 (25)
Fatigue	42 (20)	98 (22)
Diarrhea	32 (15)	71 (16)
Nausea	22 (11)	53 (12)
Grade 3/4 laboratory abnormalities		
Hemoglobin (< 90 g/L)	1 (0.5)	0
Absolute neutrophils (< 0.75 x 10^9 cells/L)	6 (3)	3 (1)
Lymphocytes (< 0.5 x 10^9 cells/L)	5 (2)	3 (1)
Platelets (< 50 x 10^9 cells/L)	9 (4)	2 (0.5)
ALT (> 5 x ULN)	3 (1)	12 (3)
AST (> 5 x ULN)	3 (1)	9 (2)
Total bilirubin (> 2.5 x ULN)	2 (1)	1 (0.2)

AE, adverse event; ALT, alanine aminotransferase; ULN, upper limit of normal; AST, aspartate aminotransferase.
[a]Includes DCV + ASV-treated patients only.
[b]Most commonly ALT/AST elevations, which resolved off-treatment (7 patients; 6/7 including patient with cirrhosis achieved SVR_{12}).

- DCV + ASV was well tolerated in patients with cirrhosis, with no clinically meaningful differences in safety parameters in cirrhotic versus non-cirrhotic patients
- Comparison of DCV + ASV versus placebo in the initial 12 weeks in treatment-naive patients with cirrhosis showed no serious adverse events, adverse events leading to discontinuation, or grade 3/4 hematologic abnormalities or aminotransferase/total bilirubin elevations with either treatment

SUMMARY

- In this global, phase 3 study with a high proportion of cirrhotic patients (≈ 30%), all-oral DCV + ASV achieved high SVR_{12} rates
 - SVR_{12} achieved in 84% and 85% of cirrhotic and non-cirrhotic GT 1b patients, respectively
 - In cirrhotic patients, SVR_{12} rates were 91% in treatment-naive, 87% in nonresponder, and 81% in IFN-ineligible/intolerant groups
- DCV + ASV was generally well tolerated
 - No clinically meaningful differences between cirrhotic and non-cirrhotic patients in safety parameters
- DCV is being further evaluated in all-oral combination regimens in multiple patient populations of high unmet need

The International Liver Congress™ 2014: The 49th Annual Meeting of the European Association for the Study of the Liver. London, UK, April 9–13, 2014